A Phase I Study of VB4-845 in Patients with Advanced, Recurrent Head and Neck Cancer on a Weekly Dosing

Abstract #5569

D. Fitsialos[1], J. Quenneville[1], M. Rasamoelisolo[2], M. Cross[1], N. Glover[1], G. MacDonald[2], Prof. Dr. M.H.H. Federico[3], Prof. Dr. C.H.E. Barrios[4], Dr. R.C. Guimarães[5], Dr. U.R. Nicolau[6]

Viventia Biotech Inc., Toronto, ON[1], Viventia Biotech Inc., Winnipeg, MB[2], Hospital das Clínicas da Faculdade de Medicina da Universidade de São Paulo[3], Hospital São Lucas – PUCRS[4], Biocor – Hospital de Doenças Cardiovasculares Ltda[5], Hospital de Câncer de Barretos – Fundação Pio XII[6]

VIVENTIA BIOTECH INC.

Updated Abstract

Background: VB4-845 (Proxinium™) is a recombinant protein engineered from the fusion of a humanized scFv specific for the epithelial cell adhesion molecule, Ep-CAM, to a truncated form of Pseudomonas Exotoxin A. Ep-CAM is highly expressed on carcinoma cells of epithelial origin, with limited normal cell expression. VB4-845 selectively targets and kills Ep-CAM-positive tumors. A phase I dose-escalation trial was completed to determine the safety, tolerability, pharmacokinetic (PK) profile and preliminary efficacy of intratumorally injected VB4-845 in patients with advanced Squamous Cell Carcinoma of the Head and Neck (SCCHN).

Methods: Dose escalation proceeded using a modified Fibonacci dose escalation scheme with a minimum of 3 patients enrolled per dose level. VB4-845 was administered once per week for 4 consecutive weeks by intratumoral injection at escalating dose levels of 100, 200, 330, 500, 700 and 930 µg/day. Dose escalation could proceed once patients in the previous dose cohort had completed 2 weeks of treatment without experiencing dose limiting toxicity. The lesion of longest diameter and suitable for accurate repeated measurements was injected with VB4-845. All toxicities were assessed according to the NCI Common Toxicity Criteria. Blood samples were collected at different times in the study to determine PK and to assess the immunogenicity of VB4-845. CT scans and tumor volume measurements where analyzed in order to explore the efficacy of VB4-845 administration according to this schedule.

Results: A total of 20 patients were enrolled onto this study. Dose-limiting toxicity defined as transient, reversible liver function tests determined the maximum tolerated dose (MTD) of VB4-845 to be 930 µg/day according to this dose schedule. In general, VB4-845 demonstrated a favorable safety profile with only mild to moderate treatment-related side effects observed. The half-life of VB4-845 was determined to be approximately 5 hours with most patients developing a systemic anti-ETA response by Day 15 of dosing. Independently-verified objective clinical responses were observed in 63% of Ep-CAM-positive patients. Four patients with Ep-CAM-positive tumors (25%) showed a complete response. The overall tumor growth control rate (objective responses plus stable disease) for Ep-CAM positive patients was 88%.

Conclusions: VB4-845 administered on a weekly basis for 4 weeks was safe and well tolerated. The MTD on this schedule was defined at 930 µg/day. Moreover, although this study was primarily designed to evaluate safety and tolerability, clinical observations indicate that VB4-845 demonstrated promising anti-tumor responses against Ep-CAM-positive SCCHN tumors in a highly treatment-refractory patient population. Further development is therefore warranted.

Background

- Head and neck cancer is the sixth most common cancer (~5% of newly diagnosed malignancies in North America)
- VB4-845 is a recombinant protein engineered from the fusion of a humanized scFv antibody fragment specific for the epithelial cell adhesion molecule, Ep-CAM, to a truncated form of Pseudomonas Exotoxin A (ETA)
- Ep-CAM is highly expressed on carcinoma cells of epithelial origin including Squamous Cell Carcinoma of the Head and Neck, and has limited normal cell expression
- Mechanism of action:
 - VB4-845 selectively binds to Ep-CAM-positive tumors and is internalized by the tumor cell through an endocytic pathway
 - The ETA portion of VB4-845 is then:
 - Efficiently dissociated within the endosome
 - Trafficked to the endoplasmic reticulum
 - Shuts off protein synthesis via binding to ribosomal eF2
 - Results in tumor cell death via apoptosis
- A phase I trial was completed to determine the maximum tolerated dose (MTD) and the PK profile of intratumorally injected VB4-845 in patients with advanced squamous cell carcinoma of the head and neck (SCCHN)

Objectives

Primary
- To determine the maximal tolerated dose (MTD) of intratumorally injected VB4-845 when injected once a week for 4 weeks

Secondary
- To determine the safety and feasibility of intratumorally injected VB4-845
- To evaluate the tumor-response of intratumorally injected VB4-845
- To evaluate the pharmacokinetic profile of intratumorally injected VB4-845

Eligibility

Principal Inclusion Criteria
- Histologically confirmed diagnosis of advanced squamous cell carcinoma of the head and neck (SCCHN) that has recurred after radiotherapy and/or chemotherapy
- Measurable disease of at least one amenable target lesion
- Age ≥ 18 years, signed informed consent
- Karnofsky performance status of ≥ 70
- Estimated life expectancy > 3 months
- Adequate hepatic, hematological and renal function

Principal Exclusion Criteria
- Clinical evidence of brain metastases
- Suspected or impeding airway obstruction
- Suspected or documented carotid artery infiltration of the lesion to be injected
- HIV, Hepatitis C (HCV), Hepatitis B
- Treatment with any other monoclonal antibody or investigational product within 4 weeks prior to screening visit
- Uncontrolled hypertension
- Radiation therapy or systemic corticosteroids within 4 weeks prior to screening visit

Trial Design

Phase I, open-label, single arm, safety and tolerability study in ascending dose cohorts with a modified Fibonacci dose escalation scheme

- 7 planned dose cohorts: 100 µg, 200 µg, 330 µg, 500 µg, 700 µg, 930 µg and 1240 µg per day with at least 3 patients per dose cohort
- 20 patients enrolled in 8 months

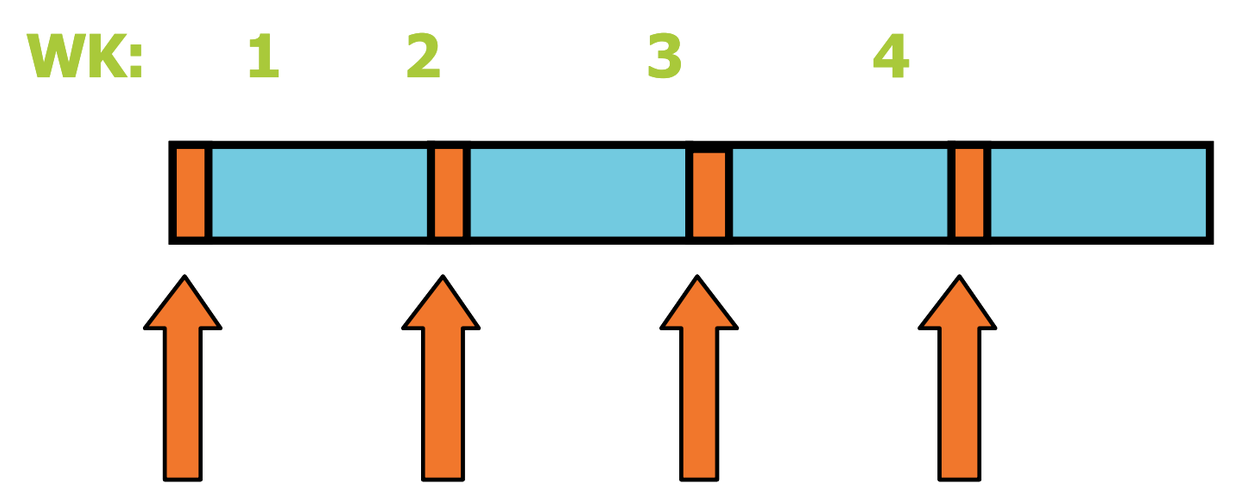
WK: 1 2 3 4

- Weekly dosing for 4 weeks
- PK assessments on Day 1 (pre-dose, 10 min, 30 min, then at 1,2,4,6,12 and 24 hrs following first dose) and a single pre-dose sample on Days 8, 15 and 22 and 4 weeks post dosing
- Anti-VB4-845 and neutralizing antibodies on Days 1, 8, 15 and 22 (pre-dosing) and 4 and 8 weeks post last dose
- Safety assessed from AEs, lab tests, physical examination, vital signs, chest X-Ray, ECG, pain assessment
- Objective tumor responses assessed from tumor measurements by caliper, CT Scans and digital photography
- Dose escalation decision after review of safety and toxicity after 2 weeks of treatment of each dose cohort

Definition of Maximum Tolerated Dose (MTD)
- The dose at which 2 patients experience a dose-limiting toxicity (DLT) as evaluated within their first 2 doses of treatment

A DLT was defined as any drug-related:
- Grade 4 toxicity for local reaction due to intratumoral injection
- Grade 4 flu-like symptom
- Grade 4 hematological toxicity
- Any other grade 3 non-hematological toxicity

Patient Characteristics (N=20)

Age– median (range)	52 years (43-77)
Gender	
Male	18
Female	2
Race	
Black	5
Caucasian	14
Other	1
Karnofsky Performance Status	
70	5
80	7
90	6
100	2
Prior Therapy	
Chemotherapy	18
1 Cycle	6
2 Cycles	9
3 Cycles	3
Radiotherapy	20

Treatment Summary

Dose (g/day)*	N	DLTs
100	3	0
200	3	0
330	3	0
500	3	0
700	3	0
930	5	2

* Administered weekly for 4 weeks.
The Dose Limiting Toxicities were transient and reversible elevated liver function tests.
The Maximal Tolerated Dose was reached at 930 µg/day.

Most Common Adverse Events

Most Common Adverse Events (number of patients experiencing event)	Number of patients per grade* Grade 1	Grade 2	Grade 3	Grade 4
Injection site reaction (pain, edema, erythema	4	2	5	0
Tumor pain	3	6	8	0
GI disorders (nausea, vomiting, loss of appetite)	7	5	1	0
Elevated liver function tests	0	1	3	1
Tumor bleeding	1	0	1	1
Dehydration	2	4	0	0
Fever	8	2	0	0

* In the situation where a subject may have had an AE more than once, the AE with the highest grade level was accounted for.

Adverse Events Related to Study Drug

Number of patients with Adverse Events Related to Study Drug (N=16)

Adverse event (number of patients experiencing event)	Number of patients per grade* Grade 1	Grade 2	Grade 3	Grade 4
Injection site reaction (pain, edema, erythema)	4	2	5	0
Tumor pain	1	0	1	0
GI disorders (nausea, vomiting, loss of appetite)	0	3	1	0
Elevated Liver function tests	0	0	3	1
Fever	5	0	0	0

* In the situation where a subject may have had an AE more than once, the AE with the highest grade level was accounted for.

Pharmacokinetics

Dose Level (g)	# of Patients[1]	Cmax (pg/mL)	Tmax (h)	$AUC_{0\text{-}last}$ (pg/mL*h)	$T_{1/2}$ (h)
100	1	1145	0.16	1955	3.38
200	2	2090	0.16	6915	3.85
330	1	1269	0.16	1886	1.84
500	0	ND	ND	ND	ND
700	2	10936	0.75	38802	4.45
930	4	14997	1.58	32384	5.95

VB4-845 plasma concentration was measured using an MTS-based assay to determine cytoxicity against the SCCHN cell line Cal-27. [1]Indicates number of patients in each dose level that had a detectable level of VB4-845 . Where >1 patient at a given dose level had detectable VB4-845 in the plasma, the values reported are the mean. ND, not determined.
- Only 10 of the 19 patients tested had detectable levels of VB4-845 in the plasma.
- 8 of the 9 patients with undetectable levels of VB4-845 had a HAPA response at time 0
- Mean Cmax (peak exposure) varied from 1145 at the lowest dose to 14997 at the highest dose with the corresponding Tmax ranging from 10min to 1.58hr.
- The overall mean extent of exposure (AUC0-last) ranged from 1955 to 38802(pg/mL)
- The mean half-life (T1/2) of VB4-845 at each dose level ranged from 2.9 to 5.1 hours with a mean half-life across all doses being 4.7±2.5 hrs.

Immunogenicity of Proxinium: HAPA and HAHA responses

Day	HAPA # Patients	HAPA Titer Range ($x10^{-3}$)	HAHA # Patients	HAHA Titer Range ($x10^{-3}$)
0	15/19	1 - 8	6/19	1
8	7/19	1-1 - 26.3	0/19	<1
15	13/18	4.9 - $1.05x10^3$	11/18	1.45 - 14
22	16/18	7.9 - $1.41x10^3$	15/18	2.2 – 129
28	17/17	3.5 – $1.31\ x10^3$	15/17	3.0 – 118
56	11/11	11.5 – $1.51\ x10^3$	9/11	4.2 - 38

Plasma samples were collected at different time points and assessed for HAPA and HAHA responses using an ELISA-based assay. Antibody titer is defined as the reciprocal of the greatest dilution of sera giving a positive OD (OD blank+4SD).
- 15 of 19 patients tested had measurable pre-bleed antibody titers to Pseudomonas exotoxin A (anti-PE or HAPA response)
- All patients tested had developed a HAPA response by day 28
- All but 2 patients eventually developed a HAHA response by the end of the study but the titer was much lower than the HAPA response

Assay for Neutralizing antibodies

Pt	Dose Level (µg)	Increase in viable cells (%) (Pre-treatment)[1]	(Post-treatment)[2]
1	100	3%	6%
3	100	4%	13%
6	200	7%	89%
7	330	18%	43%
8	330	4%	0%
10	500	0%	72%
11	500	3%	96%
18	930	0%	72%
19	930	3%	0%

A MTS-based assay was used to detect the presence of neutralizing antibodies as indicated by their ability to inhibit VB4-845 cytotoxicity. 1The neutralizing assay was done in presence of 0.1ng/mL of VB4-845 and 2 in presence of 10ng/mL of VB4-845. Shaded patient #'s indicates pre-sensitized to PE.
- A weak neutralizing antibody subset was detected in the pre-bleed samples (Time 0) in 7 patients, all of which had been pre-sensitized to PE
- By the end of the study 7 patients exhibited strong neutralizing antibody effects against VB4-845, only 2 of these were not detected in the pre-bleed sample (Time 0)
- The likelihood of developing systemic neutralizing antibodies was independent of the amount of VB4-845 dosed

Summary of Response Data By Ep-CAM Positivity

	Complete Response	Response	Stable	Progression	Non-evaluable	Total
Total number of patients EP-CAM positive (N)	4	6	4	2	2	18
Percentage Ep-CAM positive (%)	22.2	33.3	22.2	11.1	11.1	100
Percentage Ep-CAM positive and Evaluable (%)	25	37.5	25	12.5	-	100
Total number of patients Ep-CAM negative (N)	0	0	1	1	0	2
Percentage Ep-CAM negative (%)	0	0	0	50	0	100
Percentage Ep-CAM negative and Evaluable	0	0	50	50	-	100

Overall Response in Evaluable Ep-CAM positive patients:
- 63% Objective Response
- 88% Tumor Growth Control Rate (objective response + stable disease)

Complete responses: Examples

Pt 7: 4 doses of 330 µg/day

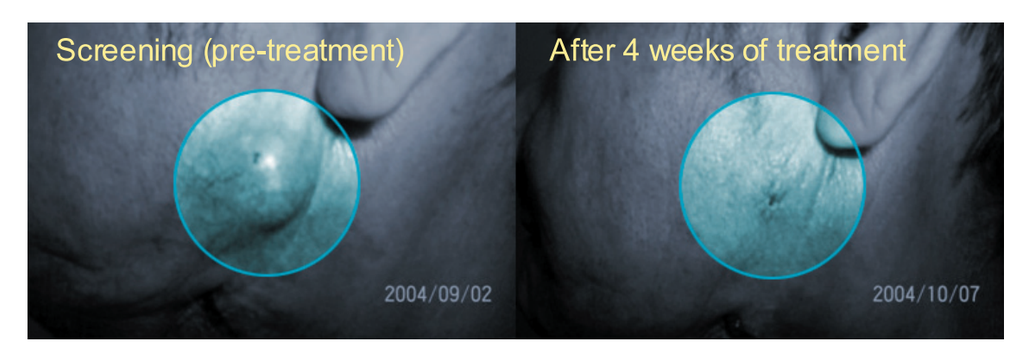
Screening (pre-treatment)
After 4 weeks of treatment
2004/09/02
2004/10/07

Pt 19: 4 doses of 930 µg/day

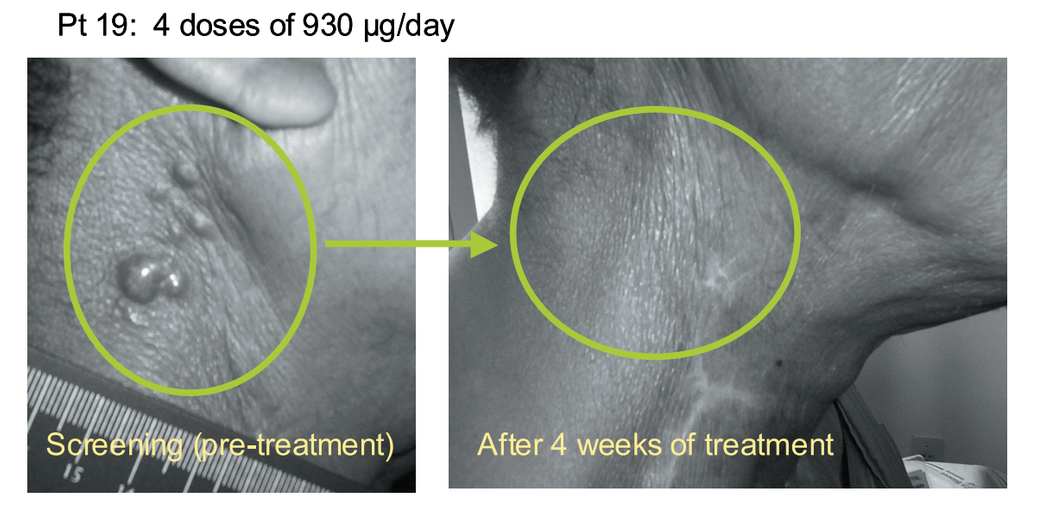
Screening (pre-treatment)
After 4 weeks of treatment

Survival:

Median overall survival data is not as yet established. The ongoing data show that:
- **6 out of 14 evaluable (133+ - 245+ days) Ep-CAM positive patients alive (including 3 patients who had CR's)**
- **No Ep-CAM negative patients still alive**

Summary and Conclusions:

- VB4-845, a recombinant fusion protein specific to Ep-CAM, can be safely and repeatedly administered to patients with advanced Head and Neck Squamous Cell Carcinoma via intratumoral injection, on a weekly schedule of administration
- The MTD for weekly dosing was established as 930 µg/day with the dose-limiting toxicity being transient and reversible liver function tests
- A safe and tolerable recommended dose of 700 µg/day was established
- Half-life of VB4-845 was approximately 5 hrs
- Systemic anti-drug antibodies are produced within 15 days of drug administration
- Although not designed to establish efficacy, this study demonstrated:
 - a 63% objective response rate with
 - a 88% overall tumor growth control rate in this advanced, therapy-resistant patient population
 - and a 25% of evaluable Ep-CAM positive patients achieved a complete response
- These promising safety and efficacy results support further clinical development